Modeling of Chemotherphy Induced Febrile Neutropenia Using the Predicted Degree and Duration of Myelosuppression

UPPSALA UNIVERSITET

Emma K. Hansson[1], Marie Sandström[2], Henrik Lindman[3] and Lena E. Friberg[1]

[1]Division of Pharmacokinetics & Drug Therapy, Uppsala University, Sweden, [2]Astra Zeneca R&D, Södertälje, Sweden,
[3]Department of Oncology, Radiology and Clinical Immunology, Uppsala University Hospital, Uppsala, Sweden

Introduction

Chemotherapy-induced neutropenic episodes are associated with the risk of developing the potentially life-threatening condition febrile neutropenia (FN) (fever ≥ 38.3 °C, absolute neutrophil count (ANC) ≤ 0.5 x 10^9/L).

FN is associated with substantially morbidity and mortality, high treatment costs, impairment in the patients quality of life and treatment disruptions.

Objectives

To develop a pharmacokinetic-pharmacodynamic model describing the time course of neutrophils in breast cancer patients either treated with epirubicin and docetaxel (ET), or 5-fluorouracil, epirubicin, and cyclophoshamide (FEC).

To characterize the relationship between FN and predicted summary measures of the myelosuppression profile as proposed by Minami [1] (Figure 1).

- a line (the duration of neutropenia)
- the area of the neutrophil-time course below a certain value
- the absolute nadir

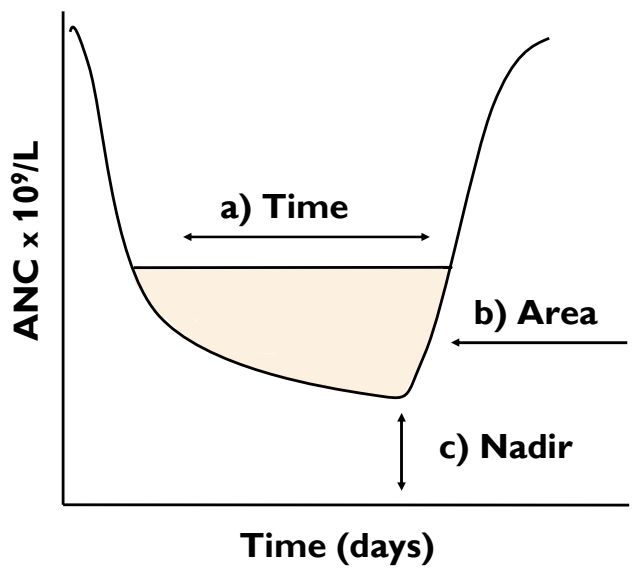
ANC x 10^9/L
a) Time
b) Area
c) Nadir
Time (days)

Figure 1. Parameters related to FN
a) Time - duration of neutropenia,
b) Area - the area of the neutrophil-time course below a certain value, c) Nadir - a single observation point

Methods

The analysis included 1053 neutrophil observations and 11 episodes of FN in 40 patients treated with ET and 1171 neutrophil observations and 8 episodes of FN in 60 patients treated with FEC.

Individual concentration-time profiles of the drugs were predicted from previous models [2, 3] and the time course of myelosuppression was described using a semi-physiological model [4]. Figure 2.

The PRIOR option in NONMEM VI was used to estimate separate potency parameters for the co-administered drugs.

Each of the model-predicted myelosuppression variables (nadir, duration or area of grade 3 or 4 neutropenia) were related to the FN data in a logistic regression model.

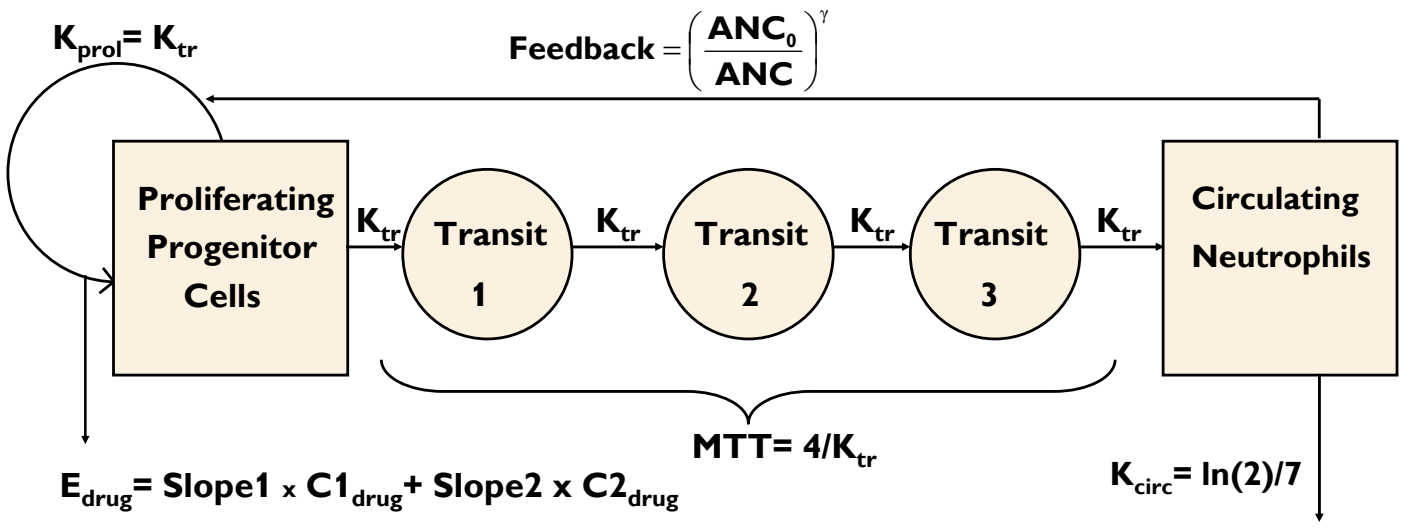
K_{prol}= K_{tr}
Feedback = $\left(\frac{ANC_0}{ANC}\right)^{\gamma}$
Proliferating Progenitor Cells
K_{tr}
Transit 1
Transit 2
Transit 3
Circulating Neutrophils
MTT= 4/K_{tr}
E_{drug}= Slope1 x $C1_{drug}$+ Slope2 x $C2_{drug}$
K_{circ}= ln(2)/7

Figure 2. The Semi-physiological model of myelosuppression.
The model consist of one compartment representing the proliferating cell pool, three transit compartments with maturating cells and one compartment of circulating observed neutrophils. MTT is the mean transit time though the chain, K_{tr}; rate constants, E_{drug}; drug effect proportional to the drug concentration and Feedback; a feedback loop from the circulating cells.

Results

The myelosuppression model could well characterize the neutrophil-time course following ET and FEC treatment and resulted in similar system-related parameter estimates as for other drugs [4] (Table 1). The fit improved when using a fixed neutrophil half-life of 7 hours and Box-Cox transforming the data with a factor of 0.2.

Table 1. *Final population model parameter estimates for FEC and ET semi-physiological myelosuppression model*

Parameter	ET		FEC	
	Estimate	IIV (%)	Estimate	IIV (%)
ANC_0 (x 10^9/L)	4.02	35	4.58	23
MTT (h)	116		185	
γ	0.18		0.24	
Slope $_{Epirubicin}$ (L/mg)	14.8	20	16.7	27.1
Slope $_{Docetaxel}$ (L/mg)	40.1	20		
Slope $_{4\text{-}OHCP}$ (L/mg)			31.2	27.1
Residual error *	0.55		0.58	

Slope $_{5\text{-}fluoruracil}$ was not significantly different from zero
* Additive residual error on Box-Cox scale

Of the investigated variables duration of grade 4 neutropenia (ANC ≤ 0.5 x 109/L) with pre-treatment neutrophil count as a covariate was best related to FN (Eq.1, 2).

$$E = \frac{E_{max} \cdot T_4^{\gamma}}{T_4^{\gamma} + T_{450}^{\gamma}} \quad \text{Eq.(1)}$$

$$PFN = \frac{EXP(E + B_{FN} + \theta \cdot ANC_0)}{1 + EXP(E + B_{FN} + \theta \cdot ANC_0)} \quad \text{Eq.(2)}$$

Eq.1, 2. *T_4; Time of grade 4 neutropenia, T_{450}; time of grade 4 neutropenia that produces 50 % of the maximal effect, PFN; Probability of developing FN, B_{FN}; Baseline probability of developing FN, ANC_0; pre-treatment neutrophil count*

Conclusion

Both the neutropenia duration and pre-treatment neutrophil count were related to the development of FN. To be able to a priori predict which patients who are at risk of developing FN, information on previous treatment and patient characteristics will be evaluated, as well as other types of models, e.g. time to event models.

References

[1] Minami H.J Clin Oncol 23:405-406, 2005
[2] Sandström M, et al. Cancer Chemother Pharmacol 58:143-156, 2006
[3] Sandström M, et al. J Clin Oncol 23: 4173-4721, 2005
[4] Friberg L, et al. J Clin Oncol 20: 4173-4721 2002